#1061

Effectiveness outcomes by treatment-free interval among patients receiving palbociclib plus endocrine therapy in HR+/HER2- advanced breast cancer in the real-world setting: Results from the PERFORM study

Bartsch, Rupert[1]; Korell, Matthias[2]; Pfeiler, Georg[3]; Gabrysiak, Thomas[4]; Fietz, Thomas[5]; Deryal, Mustafa[6]; Petersen, Volker[7]; Resch, Thomas[8]; Schöttker, Björn[9]; Decker, Thomas[10]; Müller, Lothar[11]; Radosa, Julia[12]; Rhein, Uwe[13]; Glasstetter, Martin[14]; Klein, Dunja[14]; Buder, Anna[15]; Kruger, Hagen[16]; Quartier, Azita[17]; Dzieran, Johanna[16]; Lux, Michael Patrick[18]

1. Medical University of Vienna, Department of Medicine I, Division of Oncology, Vienna, Austria | 2. Johanna Etienne Hospital, Department for Gynecology, Neuss, Germany | 3. Medical University of Vienna, Division of General Gynecology and Gynecologic Oncology, Vienna, Austria | 4. Specialized Practice for Hematology and Internal Oncology, Wolfsburg, Germany | 5. Specialized Practice for Hematology, Oncology, and Gastroenterology, Singen, Germany | 6. Caritas Hospital Saarbrücken, Center for Women's Health, Saarbrücken, Germany | 7. Oncology Practice Heidenheim, Heidenheim a.d.B, Germany | 8. Specialized Practice for Gynecology and Obstetrics, Brandenburg a.d. Havel, Germany | 9. Hematology-Oncology Practice Würzburg, Würzburg, Germany | 10. Medicine Center for Hematology and Oncology Ravensburg, Ravensburg, Germany | 11. Oncology Practice Leer, Leer, Germany | 12. Saarland University Hospital, Department for Gynecology and Obstetrics, Homburg (Saar), Germany | 13. Central Hospital Suhl, Gynecology and Obstetrics, Suhl, Germany | 14. iOMEDICO, Freiburg, Germany | 15. Pfizer Corporation Austria GmbH, Vienna, Austria | 16. Pfizer Pharma GmbH, Berlin, Germany | 17. Pfizer PIO., Paris, France | 18. Department for Gynecology and Obstetrics, Frauenklinik St. Louise, Paderborn, Frauenklinik St. Josefs, Salzkotten, St. Vincenz Kliniken Salzkotten + Paderborn, Germany

Background

CDK4/6 inhibitors (CDK4/6i) plus endocrine therapy (ET) are first-line (1L) standard of care for patients with HR+/HER2- advanced breast cancer (ABC) based upon results of the pivotal trials[1–5]. Treatment-free interval (TFI) is a prognostic factor in ABC; a short TFI, a surrogate of endocrine resistance, is associated with more aggressive disease and poorer treatment outcomes[6,7]. Real-world (RW) evidence complements clinical trials by providing insights into patients underrepresented in clinical trials. Here we present interim analysis 5 (IA5) of the prospective non-interventional German and Austrian PERFORM study, conducted five years after first patient inclusion, describing real-world characteristics, treatment patterns, and outcomes in patients receiving 1L palbociclib + ET, stratified by TFI from last (neo) adjuvant therapy to recurrence: ≤12 months, >12 months, and *de novo* ABC.

Methods

The primary endpoint was first line progression free survival (1L PFS), defined as time from 1L treatment initiation to progression or death. Secondary endpoints included 2L PFS (time from 2L initiation to progression or death), PFS2 (time from 1L initiation to progression or death during 2L), and overall survival (OS; time from 1L initiation to death). Patients without events were censored at last contact or initiation of the next therapy line, as applicable. Patient and tumor characteristics and treatment patterns were summarized descriptively. Time to event endpoints were estimated using Kaplan-Meier methods, with observation period estimated using reverse Kaplan-Meier methodology. Tumor assessments followed local clinical practice. First line endocrine based palbociclib was defined from initiation of palbociclib or endocrine therapy to discontinuation of combination therapy; endocrine backbone switches were not considered new therapy lines unless due to disease progression.

Patient reported outcomes (PROs) were assessed using FACT-B at baseline and every 3 months during palbociclib treatment. The PRO analysis set included patients with evaluable baseline and post baseline FACT-B scores. Based on an exploratory IA5 analysis, only change from baseline during 1L treatment was evaluated. Clinically meaningful change was defined using established minimally important differences (MID)[8].

Results

PATIENT POPULATION

Between October 27th, 2020, and database cut on September 30th, 2025, 1,436 patients were enrolled at 188 study sites across Germany and Austria; all qualified for IA5 (≥6 months follow-up). Of 1,389 treated patients, 121 were excluded from the full analysis set (FAS, n = 1,268) due to off-label use (n = 52), eligibility violation of eligibility criteria identified after treatment start (n = 67) or missing palbociclib/ET start (n = 2). At database cutoff, 850 patients discontinued 1L therapy (mainly due to disease progression; n = 523 patients). TFI was available for 1,010 of 1,268 treated patients (79.7%). Missing TFI (n = 258) was due to the recurrence date before last (neo)adjuvant dose or after inclusion. Median observation time from start of 1L treatment was 36.8 months overall (95% CI: 35.8 – 38.2) and 39.3, 37.4 and 36.4 months for TFI ≤12 months, TFI >12 months and *de novo* subgroup, respectively.

PATIENT AND TUMOR CHARACTERISTICS

Median age at 1L start was 69.6 years (FAS). Patients with a TFI ≤12 months were younger (median age of 65.4 years) than those with TFI >12 months (70.1 years) and *de novo* ABC (71.2 years); with a higher proportion of pre/perimenopausal patients (11.3% vs 3.2% and 9.5%, respectively).

Invasive breast carcinoma of no special type was the most common histology (>60% across all TFI subgroups).

Patients with a TFI ≤12 months exhibited a more aggressive disease profile (G3: 30.1%; liver metastases: 26.3%), compared with patients with TFI >12 months (17.4%; 13.4%) and those with *de novo* ABC (21.2%; 14.9%). Visceral disease at 1L start occurred in 54.9%, 51.7%, and 47.6% of patients with TFI ≤12 months, TFI >12 months and 47.6% with *de novo* ABC, respectively **(Table 1)**.

EFFECTIVENESS

Overall median PFS was 25.5 months (95% CI: 23.0 – 28.6) **(Figure 1A)** and 14.6 months (95% CI: 9.3 – 19.4), 29.2 months (95% CI: 23.3 – 33.1) and 31.5 months (95% CI: 26.5 – 36.7) for TFI ≤12 months, TFI >12 months, and *de novo* ABC, respectively **(Figure 1B)**. Median PFS2 was 36.6 months overall (95% CI: 33.0 – 40.6), and 21.2 months (95% CI: 17.4 – 37.0), 39.0 months (95% CI: 33.7 – 51.4) and 40.9 months (95% CI: 37.0 – 46.9) by the same subgroups **(Figure 1C)**. Median 2L PFS was 6.0 (95% CI: 5.3 – 7.4) overall and 5.0 (95% CI: 3.3 – 5.8), 6.0 (95% CI: 4.9 – 9.1) and 7.3 months (95% CI: 5.6 – 9.4) for TFI ≤12, TFI >12, and *de novo*, respectively **(Figure 1D)**.

Median OS results were immature for some TFI subgroups at database cut; therefore 12 and 24 month OS rates are reported **(Table 2)**. Overall, the 12 month OS rate was 90.2%, and the 24 month OS rate was 76.7%; with a trend for consistently lower OS rates in patients with TFI ≤12 months versus TFI >12 months or *de novo* ABC.

QUALITY OF LIFE (QOL)

PROs were available for 1,008 patients: 105 patients (10.4%) with TFI ≤12 months, 309 patients (30.7%) with TFI >12 months and 389 patients (38.6%) with *de novo* disease. TFI could not be derived for 205 patients (PRO set). Details for the PROs are provided in **supplemental Table 1** and 2A – C. Mean change from baseline was assessed by TFI subgroups over up to 15 months, (aligned with median PFS in TFI ≤12 months: 14.6 months). FACT-B total and FACT-G scores showed no clinically relevant decrease **(Figure 2)** and subscores were generally maintained **(Supplemental Figure 1)**; indicating QOL was overall maintained irrespective of TFI.

Limitations

Limitations include the descriptive, hypothesis-generating design and exploratory QOL analyses. The study is ongoing and some data, i.e. TFI, may be incomplete. OS requires further follow-up (67.6% censored at database cut) and questionnaire completion varied over time.

Table 1: Patient and tumor characteristics (FAS)

	Total (n = 1268)	TFI >12 months (n = 373)	TFI ≤12 months (n = 133)	*De novo* ABC (n = 504)
Age at start of 1L treatment [years]				
Median (25%/75% quantiles)	69.6 (60.0 – 77.6)	70.1 (62.7 – 77.6)	65.4 (55.0 – 74.6)	71.2 (60.3 – 78.1)
<65 years [n (%)]	471 (37.1)	117 (31.4)	66 (49.6)	176 (34.9)
65 – 74 years [n (%)]	380 (30.0)	134 (35.9)	35 (26.3)	144 (28.6)
75 – 79 years [n (%)]	207 (16.3)	61 (16.4)	17 (12.8)	100 (19.8)
≥80 years [n (%)]	210 (16.6)	61 (16.4)	15 (11.3)	84 (16.7)
Sex [n (%)]				
Female	1257 (99.1)	372 (99.7)	132 (99.2)	497 (98.6)
Male	11 (0.9)	1 (0.3)	1 (0.8)	7 (1.4)
Menopausal status [n (%)]				
Pre-/Perimenopausal	87 (6.9)	12 (3.2)	15 (11.3)	48 (9.5)
Postmenopausal	1170 (92.3)	360 (96.5)	117 (88.0)	449 (89.1)
Not derivable (i.e. male patients)	11 (0.9)	1 (0.3)	1 (0.8)	7 (1.4)
ECOG Performance Status [n (%)]				
0	535 (42.2)	156 (41.8)	59 (44.4)	209 (41.5)
1	526 (41.5)	172 (46.1)	57 (42.9)	197 (39.1)
2 – 4	168 (13.3)	40 (10.7)	13 (9.8)	79 (15.7)
Not assessed / missing	39 (3.1)	5 (1.3)	4 (3.0)	19 (3.8)
Time since initial diagnosis [years]				
Median (25%/75% quantiles)	3.0 (0.1 – 9.7)	12.4 (8.6 – 16.5)	4.2 (2.2 – 6.7)	NA
Histology of primary tumor				
Cribriform carcinoma	4 (0.3)	0 (0.0)	1 (0.8)	3 (0.6)
Invasive breast carcinoma of no special type	779 (61.4)	231 (61.9)	86 (64.7)	307 (60.9)
Invasive lobular carcinoma	280 (22.1)	66 (17.7)	33 (24.8)	130 (25.8)
Mucinous carcinoma	20 (1.6)	7 (1.9)	0 (0.0)	10 (2.0)
Tubular carcinoma	8 (0.6)	4 (1.1)	0 (0.0)	2 (0.4)
Other carcinoma	177 (14.0)	65 (17.4)	13 (9.8)	52 (10.3)
Tumor grading at initial diagnosis				
G1	86 (6.8)	28 (7.5)	5 (3.8)	27 (5.4)
G2	820 (64.7)	240 (64.3)	83 (62.4)	350 (69.4)
G3	278 (21.9)	65 (17.4)	40 (30.1)	107 (21.2)
GX[a]	84 (6.6)	40 (10.7)	5 (3.8)	20 (4.0)
Location(s) of metastatic sites present at inclusion (i.e., >10%)[b]				
Bone	802 (63.2)	233 (62.5)	78 (58.6)	336 (66.7)
Liver	202 (15.9)	50 (13.4)	35 (26.3)	75 (14.9)
Lung	277 (21.8)	84 (22.5)	22 (16.5)	122 (24.2)
Lymph nodes (distal)	103 (8.1)	32 (8.6)	14 (10.5)	44 (8.7)
Lymph nodes (regional)	119 (9.4)	25 (6.7)	12 (9.0)	60 (11.9)
Pleura	142 (11.2)	66 (17.7)	13 (9.8)	37 (7.3)
Disease site present at baseline of 1L treatment [n (%)]				
Visceral	632 (49.8)	193 (51.7)	73 (54.9)	240 (47.6)
Non-visceral only (excl. bone only)	144 (11.4)	38 (10.2)	17 (12.8)	55 (10.9)
Bone only	430 (33.9)	126 (33.8)	40 (30.1)	178 (35.3)
No metastases present at inclusion*	62 (4.9)	16 (4.3)	3 (2.3)	31 (6.2)

* Subgroup without metastases present at inclusion includes patients with locally advanced breast cancer and patients with removed metastases (surgery, radiation) after initial diagnosis.
[a] Grade cannot be assessed, as per the category documented in the EDC.
[b] Multiple answers possible. Only locations with more than 10% in one of the subgroups are displayed

Table 2: Effectiveness of treatment (FAS)

	Total (n = 1268)	TFI >12 months (n = 373)	TFI ≤12 months (n = 133)	*De novo* ABC (n = 504)
Best response [n (%)] in 1L				
CR	96 (7.6)	29 (7.8)	5 (3.8)	46 (9.1)
PR	385 (30.4)	99 (26.5)	31 (23.3)	200 (39.7)
SD ≥24 weeks	407 (32.1)	142 (38.1)	46 (34.6)	137 (27.2)
SD <24 weeks	108 (8.5)	29 (7.8)	12 (9.0)	39 (7.7)
Non-CR/Non-PD	3 (0.2)	1 (0.3)	1 (0.8)	0 (0.0)
Non-PD (acc. to PI)	5 (0.4)	1 (0.3)	0 (0.0)	3 (0.6)
PD	117 (9.2)	33 (8.8)	22 (16.5)	26 (5.2)
Missing	147 (11.6)	39 (10.5)	16 (12.0)	53 (10.5)
Overall survival				
Median [month (95% CI)]	52.8 (47.9, NA)	NA (47.6, NA)	37.4 (26.7, NA)	52.8 (45.5, NA)
12-month rate [95% CI]	90.2% (88.4, 91.7)	94.5% (91.6, 96.4)	82.1% (74.2, 87.7)	91.3% (88.5, 93.5)
24-month rate [95% CI]	76.7% (74.1, 79.1)	81.2% (76.5, 85.1)	61.8% (52.3, 69.8)	80.7% (76.7, 84.1)
Reason for end of 1L treatment [n (%)]				
(Serious) Adverse Event	157 (12.4)	42 (11.3)	16 (12.0)	72 (14.3)
Progressive disease	523 (41.2)	158 (42.4)	70 (52.6)	167 (33.1)
Other*	57 (4.5)	55 (14.7)	15 (11.3)	66 (13.1)
Lost to follow-up	64 (5.0)	NA	NA	NA
Withdrawal of informed consent by subject	41 (3.2)	NA	NA	NA
No (S)AE and no PD but switch in same substance class	7 (0.6)	NA	NA	NA
Radiotherapy	1 (0.1%)	NA	NA	NA

*Category "Other" in TFI subgroups comprises all depicted categories except "(Serious) Adverse Event" and "Progressive disease".

Figure 1: Progression-free survival in total and by TFI (FAS)

A: Progression-free survival of 1L treatment, PAL + ET (n = 1268)

B: Progression-free survival of 1L treatment by TFI

C: Progression-free survival 2 by TFI

D: 2L progression-free survival by TFI

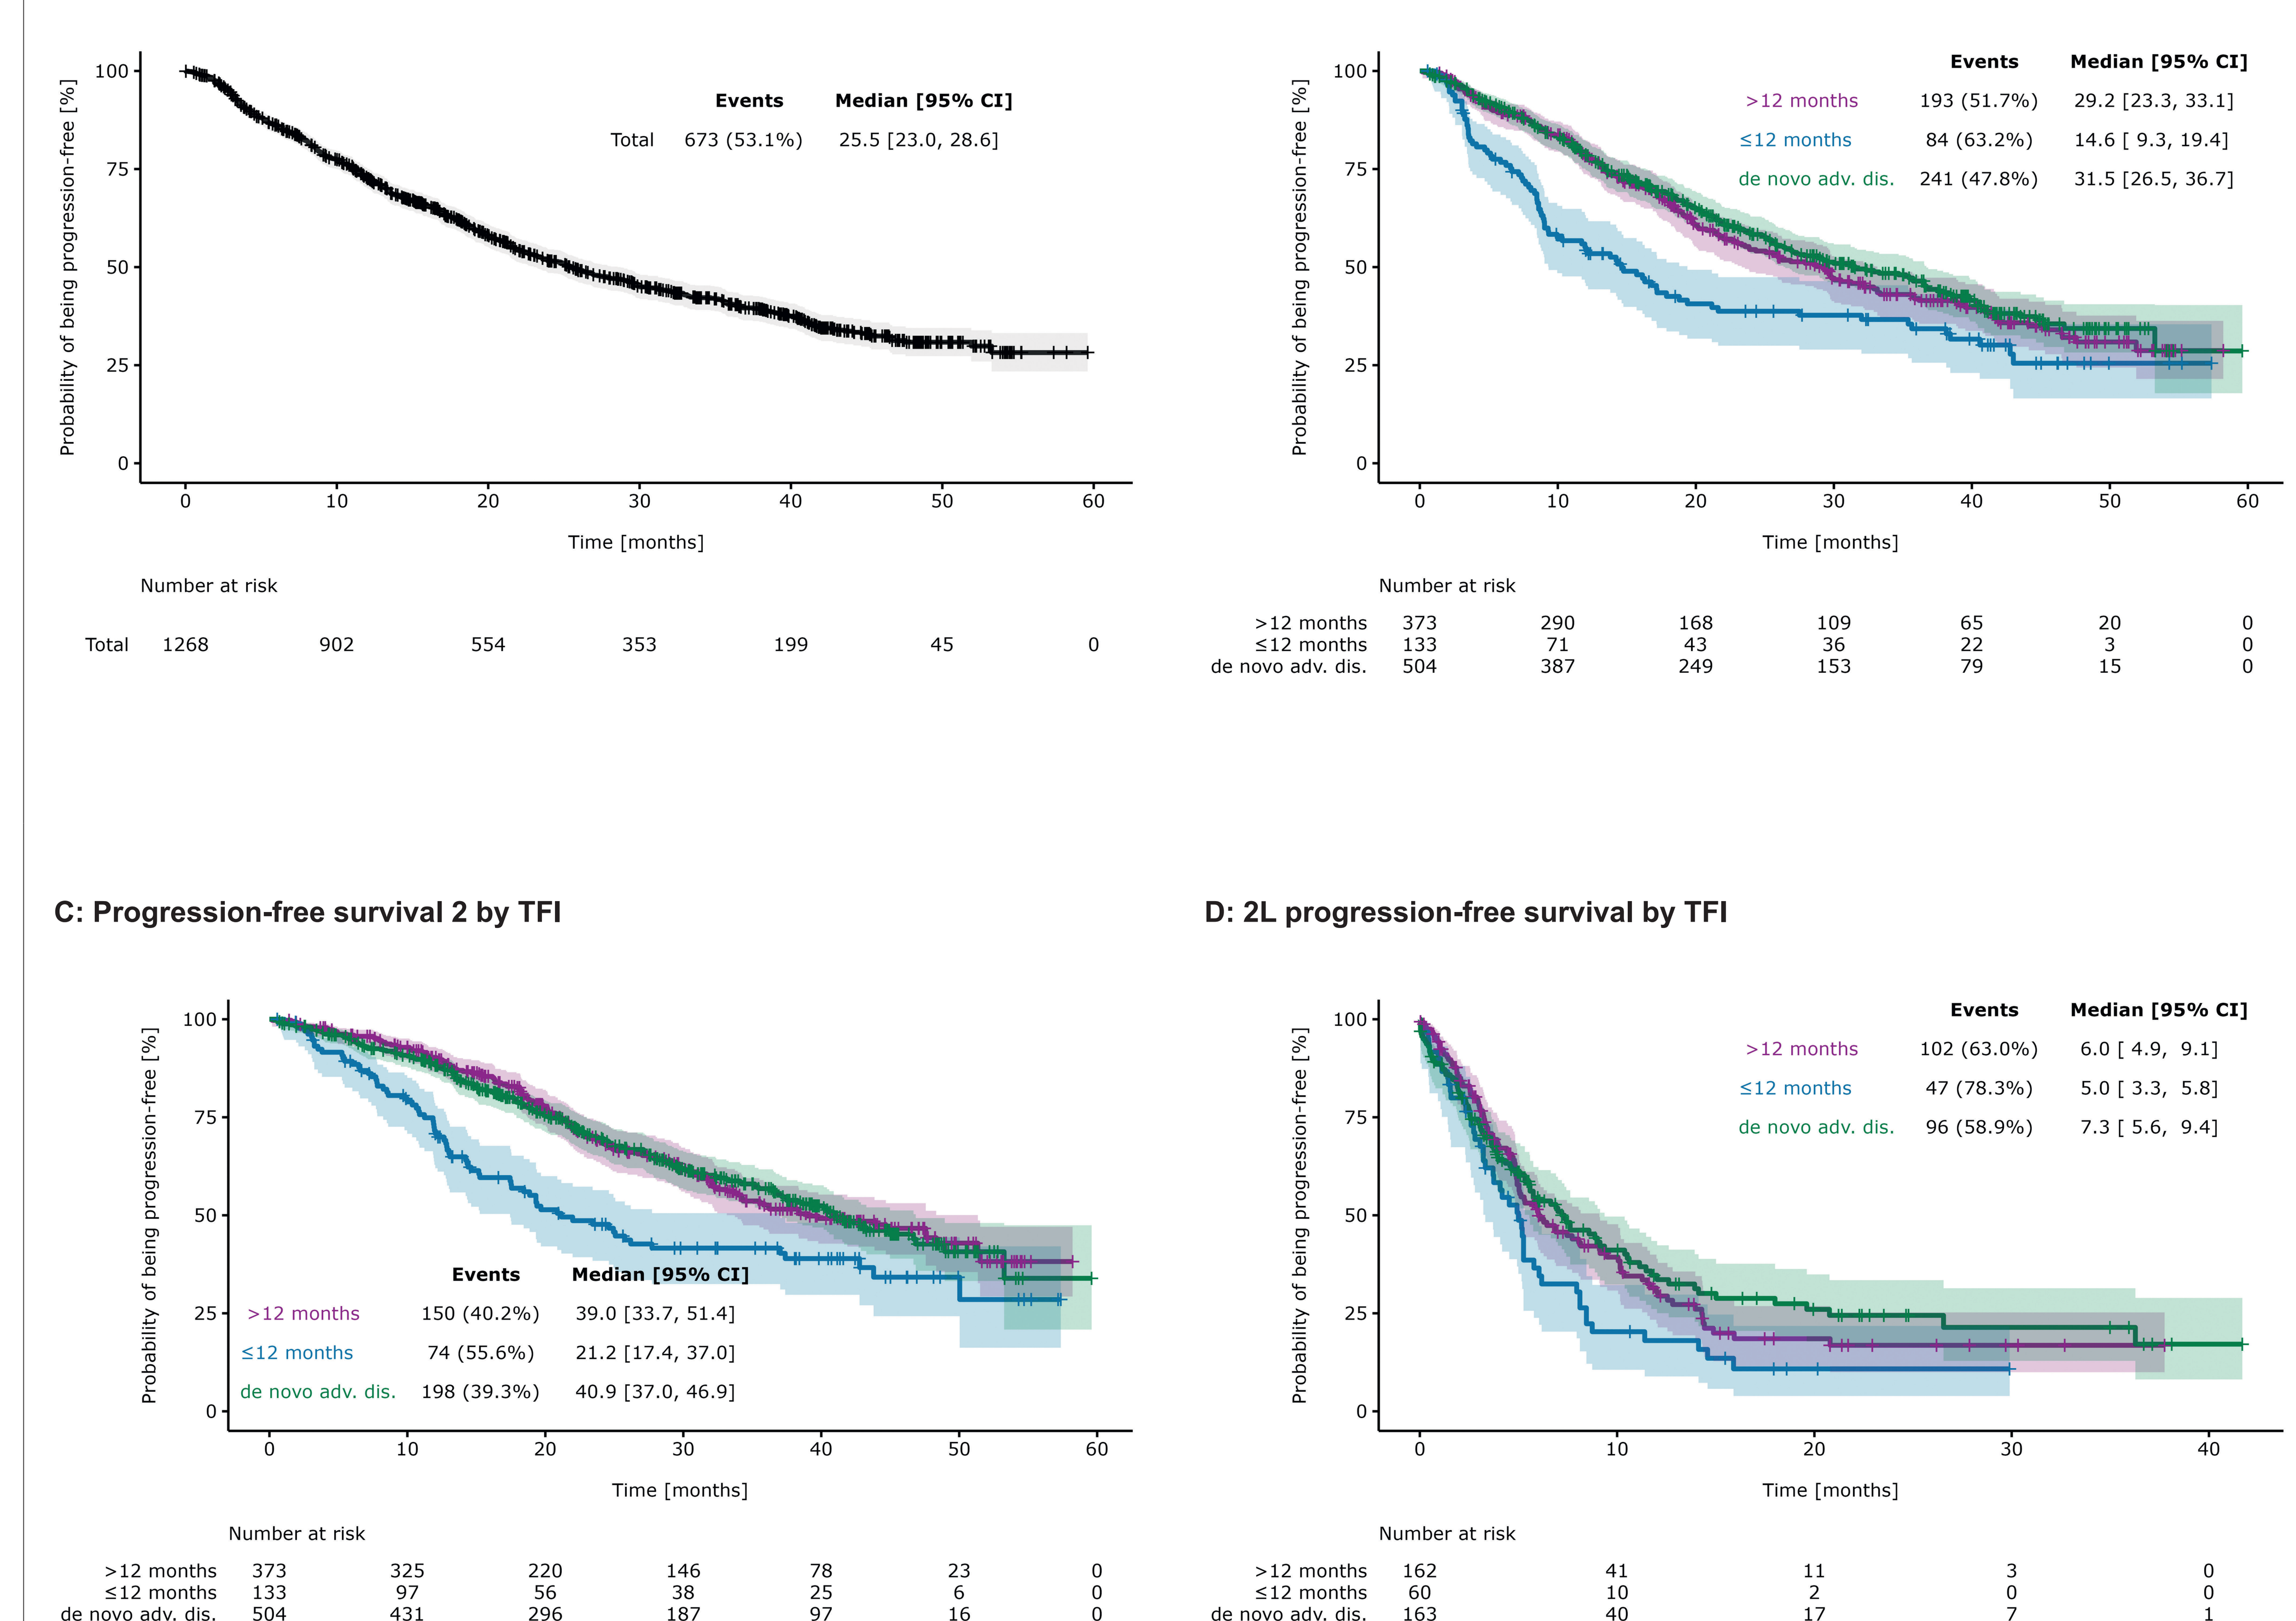
Probability of being progression-free [%]
Time [months]
Events Median [95% CI]
Total 673 (53.1%) 25.5 [23.0, 28.6]
Number at risk
Total 1268 902 554 353 199 45 0
>12 months 193 (51.7%) 29.2 [23.3, 33.1]
≤12 months 84 (63.2%) 14.6 [9.3, 19.4]
de novo adv. dis. 241 (47.8%) 31.5 [26.5, 36.7]
>12 months 373 290 168 109 65 20 0
≤12 months 133 71 43 36 22 3 0
de novo adv. dis. 504 387 249 153 79 15 0
>12 months 150 (40.2%) 39.0 [33.7, 51.4]
≤12 months 74 (55.6%) 21.2 [17.4, 37.0]
de novo adv. dis. 198 (39.3%) 40.9 [37.0, 46.9]
>12 months 373 325 220 146 78 23 0
≤12 months 133 97 56 38 25 6 0
de novo adv. dis. 504 431 296 187 97 16 0
>12 months 102 (63.0%) 6.0 [4.9, 9.1]
≤12 months 47 (78.3%) 5.0 [3.3, 5.8]
de novo adv. dis. 96 (58.9%) 7.3 [5.6, 9.4]
>12 months 162 41 11 3 0
≤12 months 60 10 2 0 0
de novo adv. dis. 163 40 17 7 1

Figure 2: Mean change from Baseline in FACT-B total score and FACT-G score by TFI during 1L treatment (PRO set)

A: FACT-B total score

B: FACT-G total score

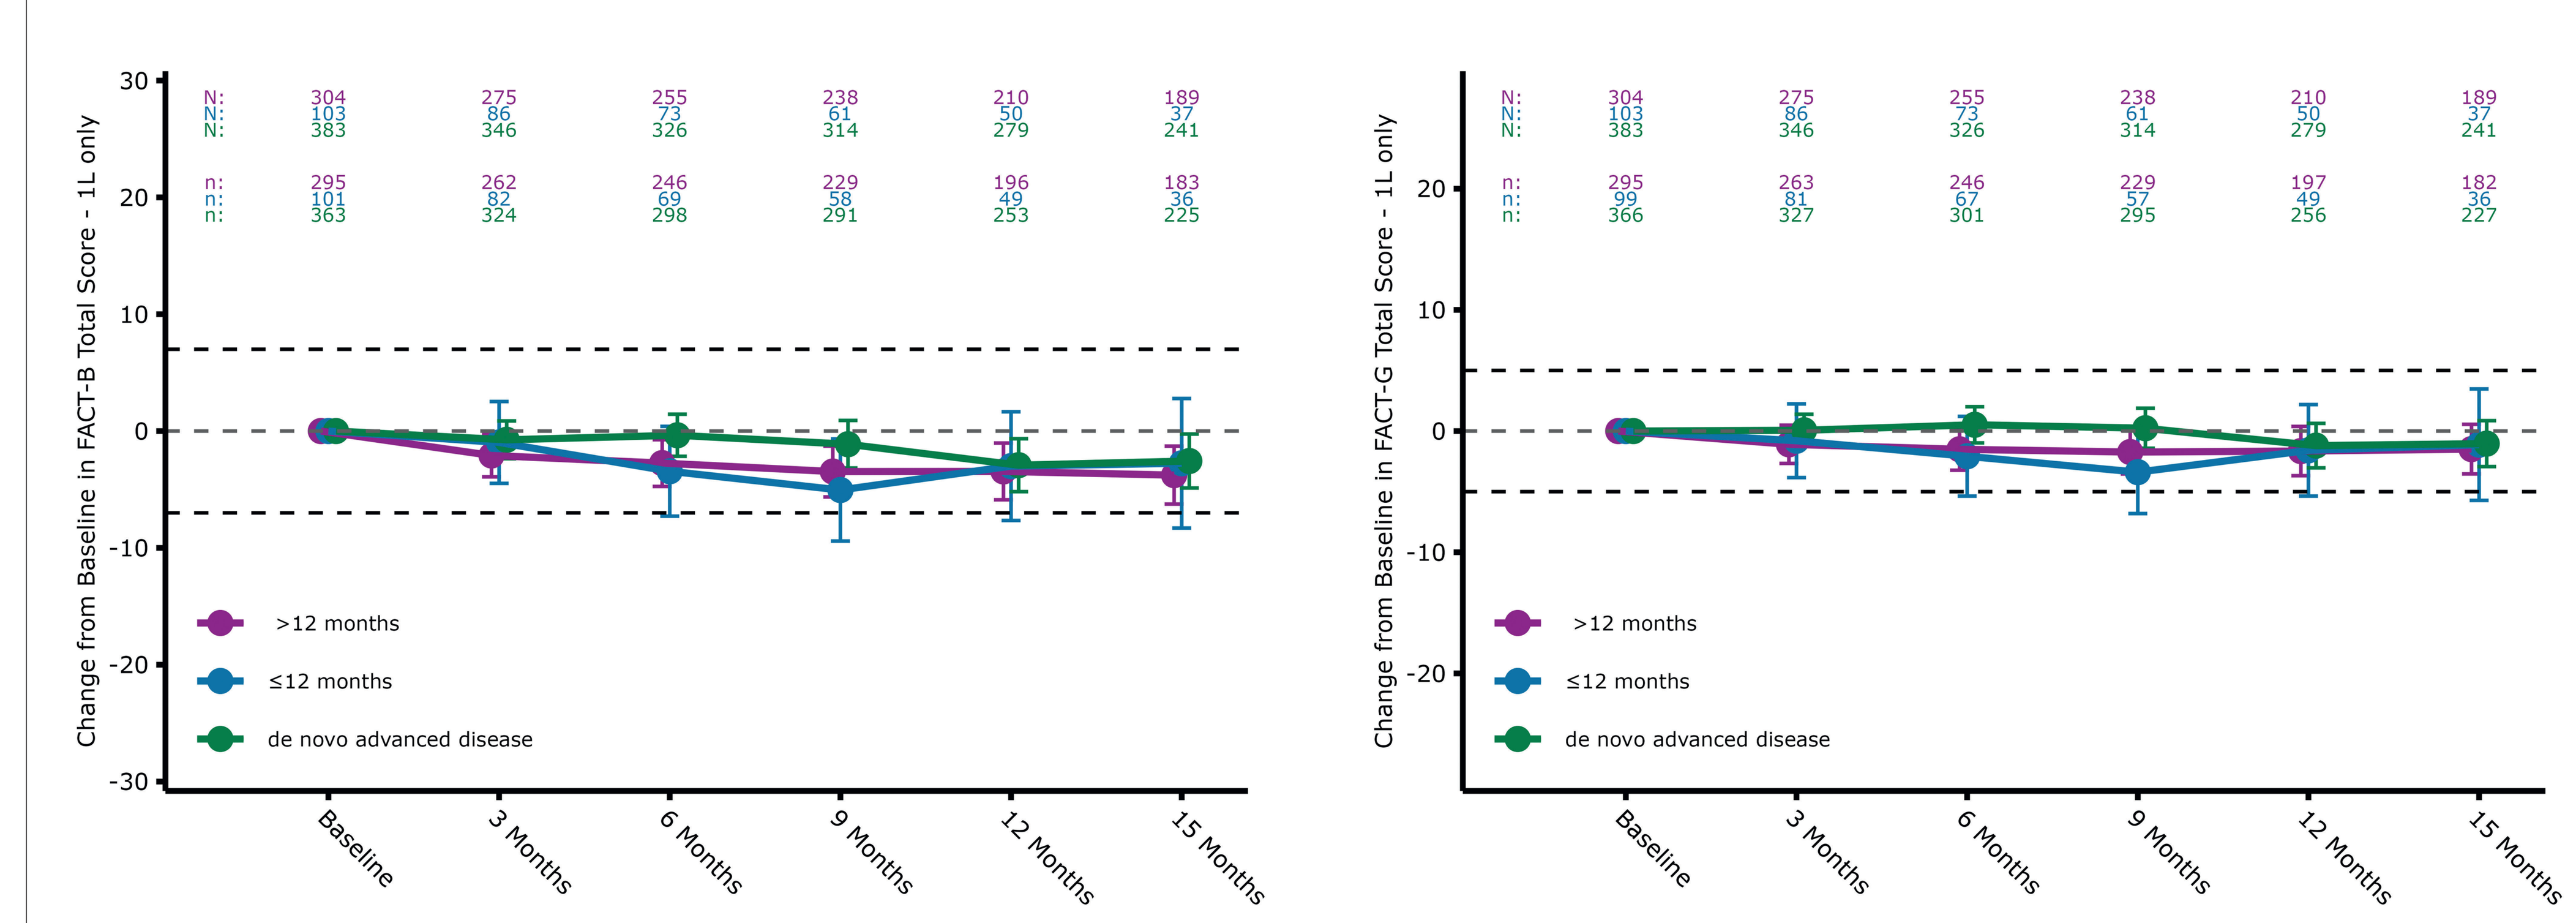
Change from Baseline in FACT-B Total Score - 1L only
Change from Baseline in FACT-G Total Score - 1L only
N: 304 275 255 238 210 189
N: 103 86 73 61 50 37
N: 383 346 326 314 279 241
n: 295 262 246 229 196 183
n: 101 82 69 58 49 36
n: 363 324 298 291 253 225
n: 295 263 246 229 197 182
n: 99 81 67 57 49 36
n: 366 327 301 295 256 227
>12 months
≤12 months
de novo advanced disease
Baseline
3 Months
6 Months
9 Months
12 Months
15 Months

Error bars indicate confidence intervals (CI) at the respective time points.
The minimally important differences (MID) in the positive and negative direction are indicated by black dotted lines.
N corresponds to the number of returned and evaluable questionnaires at the respective time point.
n corresponds to the number of evaluable responses for the respective score at the respective time point.

Discussion

PERFORM IA5 provides real-world insights into patient characteristics and effectiveness of 1L treatment with palbociclib + ET. Outcomes were consistent with and complement PALOMA-2, supporting applicability to routine clinical practice.[9] This analysis describes effectiveness and QOL by TFI.

Most patients with available TFI initiated 1L palbociclib + ET after a TFI >12 months or with *de novo* ABC. Outcomes were more favorable in patients with a TFI >12 months or *de novo* ABC than with TFI ≤12 months. This observation highlights the limited efficacy of current treatment approaches, including ET and CDK4/6i, in patients with a short TFI and underscores the need for novel therapeutic strategies in this subgroup with limited endocrine sensitivity.[10]

Patients with a TFI ≤12 months had poorer outcomes across endpoints, consistent with more aggressive disease. Given their frequent exclusion from pivotal trials, PERFORM adds valuable real-world data and highlights a substantial unmet medical need. Recent advances may be relevant: inavolisib (PI3Kα inhibitor) added to palbociclib and fulvestrant showed encouraging efficacy in PIK3CA-mutated ABC, following recurrence on or within 12 months of completing adjuvant endocrine treatment.[11] Ongoing studies targeting additional biomarkers may further expand options for patients with short TFI.

QOL data by TFI are scarce in randomized controlled trials and real world studies. In PERFORM, FACT-B total -and FACT-G changes remained within established MID thresholds for over 12 months during 1L, irrespective of TFI. This finding underscores that quality of life can be maintained during 1L treatment across TFI subgroups, including in patients who are more likely to remain on long-term therapy, such as those with a TFI >12 months or *de novo* disease.

PERFORM will continue follow-up over the next 1.5 years and will further evaluate effectiveness and QOL in additional clinically relevant subgroups.

Conclusions

In this large prospective non-interventional cohort, 1L palbociclib plus ET demonstrated clinically meaningful real-world effectiveness with QOL generally preserved. Outcomes differed by TFI, with shorter TFI associated with higher baseline risk features and shorter PFS and PFS2, while QOL was maintained across TFI subgroups. These descriptive results are consistent with prior trials and real world evidence and support TFI as clinically relevant factor to inform expectations and patient counseling.

References:
1. Finn RS, Martin M, Rugo HS et al., Palbociclib and Letrozole in Advanced Breast Cancer. N Engl J Med. 2016;375(20):1925-1936. doi:10.1056/NEJMoa1607303
2. Hortobagyi GN, Stemmer SM, Burris HA et al., Ribociclib as First-Line Therapy for HR-Positive, Advanced Breast Cancer. N Engl J Med. 2016;375(18):1738-1748. doi:10.1056/NEJMoa1609709
3. Goetz MP, Toi M, Campone M et al., MONARCH 3: Abemaciclib as Initial Therapy for Advanced Breast Cancer. J Clin Oncol. 2017;35(32):3638-3646. doi:10.1200/JCO.2017.75.6155
4. Gennari A, André F, Barrios CH, et al. ESMO Clinical Practice Guideline for the diagnosis, staging and treatment of patients with metastatic breast cancer. Ann Oncol. 2021;32(12):1475-1495. doi:10.1016/j.annonc.2021.09.019
5. Kunwor R, Baniya R, Abu-Khalaf MM. Meta-analysis of cyclin-dependent kinase (CDK) 4/6 inhibitors with endocrine therapy versus endocrine therapy alone on progression-free survival (PFS) and overall survival (OS) for metastatic breast cancer (MBC). J Clin Oncol. 2020;38(15_suppl):1060-1060. doi:10.1200/JCO.2020.38.15_suppl.1060
6. Di Leo, A., O'Shaughnessy, J., Sledge, G.W. et al. Prognostic characteristics in hormone receptor-positive advanced breast cancer and characterization of abemaciclib efficacy. npj Breast Cancer 4, 41 (2018). https://doi.org/10.1038/s41523-018-0094-2
7. Davie A, Cuyun Carter G, Rider A, Bailey A, Lewis K, Price G, Ostojic H, Ringeisen F, Pivot X. Real-world clinical profile, treatment patterns and patient-reported outcomes in a subset of HR+/HER2- advanced breast cancer patients with poor prognostic factors: data from an international study. ESMO Open. 2021 Aug;6(4):100226. doi: 10.1016/j.esmoop.2021.100226. Epub 2021 Aug 7. PMID: 34371379; PMCID: PMC8358418
8. Eton DT, Cella D, Yost KJ, Yount SE, Peterman AH, Neuberg DS, Sledge GW, Wood WC. A combination of distribution- and anchor-based approaches determined minimally important differences (MIDs) for four endpoints in a breast cancer scale. J Clin Epidemiol. 2004 Sep;57(9):898-910. doi: 10.1016/j.jclinepi.2004.01.012. PMID: 15504633.
9. Rugo HS, Finn RS, Diéras V et al., Palbociclib plus letrozole as first-line therapy in estrogen receptor-positive/human epidermal growth factor receptor 2-negative advanced breast cancer with extended follow-up. Breast Cancer Res. Treat. 174(3), 719–729 (2019).
10. Ruckhäberle E, Schmidt M, Welt A, Harbeck N, Wöckel A, Gluz O, Park-Simon TW, Untch M, Lux MP. Palbociclib: Randomized Studies and Real-world Evidence as the Basis for Therapeutic Planning in Metastatic Breast Cancer. Geburtshilfe Frauenheilkd. 2024 Sep 2;84(9):813-836.
11. Jhaveri KL, Im SA, Saura C, et al. Overall Survival with Inavolisib in PIK3CA-Mutated Advanced Breast Cancer. N Engl J Med. 2025;393(2):151-161. doi:10.1056/NEJMoa2501796

Conflicts of Interest:
Rupert Bartsch: Honoraria, Consulting or Advisory Role, Responsible Investigator Study Site, Travel/Accomodation Expenses | Matthias Korell: Responsible Investigator Study Site | Georg Pfeiler: Honoraria, Consulting or Advisory Role, Speaker's Bureau, Responsible Investigator Study Site, Travel/Accomodation Expenses | Thomas Gabrysiak: Responsible Investigator Study Site | Thomas Fietz: Consulting or Advisory Role, Responsible Investigator Study Site | Mustafa Deryal: Responsible Investigator Study Site | Volker Petersen: Responsible Investigator Study Site | Thomas Resch: Responsible Investigator Study Site, Research Funding | Björn Schöttker: Responsible Investigator Study Site | Thomas Decker: Honoraria, Consulting or Advisory Role, Speaker's Bureau, Responsible Investigator Study Site | Lothar Müller: Honoraria, Consulting or Advisory Role, Research Funding, Expert Testimony, Responsible Investigator Study Site, Travel/Accomodation Expenses | Julia Caroline Radosa: Honoraria, Consulting or Advisory Role, Expert Testimony, Responsible Investigator Study Site, Travel/Accomodation Expenses | Uwe Rhein: Responsible Investigator Study Site | Martin Glasstetter: Full/Part-time employment | Dunja Klein: Full/Part-time employment | Anna Buder: Full/Part-time employment, Stocks/Shares | Hagen, Kruger: Full/Part-time employment, Stocks/Shares | Azita Quartier: Full/Part-time employment, Stocks/Shares | Johanna Dzieran: Full/Part-time employment, Stocks/Shares | Michael Patrick Lux: Honoraria, Consulting or Advisory Role, Speaker's Bureau, Responsible Investigator Study Site, Travel/Accomodation Expenses

Acknowledgements: The non-interventional study PERFORM is sponsored by Pfizer Pharma GmbH (Berlin, Germany).

Abbreviations:
1L: First-line / First line | 2L: Second-line / Second line | ABC: Advanced Breast Cancer | Acc. to PI: According to Principal Investigator | CDK4/6i: Cyclin-Dependent-Kinase 4/6 inhibitor | CI: Confidence Interval | CR: Complete Response | ECOG: Eastern Cooperative Oncology Group Performance Status | ET: Endocrine Therapy | EWB: Emotional Well-Being | FACT-B: Functional Assessment of Cancer Therapy - Breast | FAS: Full Analysis Set | FWB: Functional Well-Being | HER2: Human-Epidermal-Growth-Factor-Receptor 2 | HR: Hormone Receptor | IA: Interim Analysis | MID: Minimally Important Difference | n: Number of patients | NA: Not Applicable | OS: Overall Survival | PD: Progressive Disease | PFS: Progression-Free Survival | PFS2: Progression-Free Survival 2 | 2L-PFS: Second-line Progression-Free Survival | PR: Partial Response | PRO: Patient-reported Outcomes | PWB: Physical Well-Being | QOL: Quality of Life | RW: Real World | (S)AE: (Serious) Adverse Event | SD: Stable Disease | SWB: Social Well-Being | TFI: Treatment-free Interval | TOI: Trial Outcome Index

Presented at ASCO congress 2026 • May 29 – June 2, 2026 • Chicago • Posternumber 1061

Copyright ©2026. All rights reserved.